LB-306

Impacting Humoral Hypercalcemia of Malignancy (HHM) and associated PTH1R-mediated morbidities: Characterization of an Anti-PTH1R antagonist monoclonal antibody to reverse hypercalcemia

Amer Mirza*, Daniel Bedinger, Rachel Hunt, Robyn Cotter, Elizabeth Pongo, Kiran Ahluwalia, Catarina Tran, Llewelyn Lao, Kristin Lind, Sujeewa Wijesuriya, Lynn Webster, Fangjiu Zhang, Agnes Choppin, Ou Li, Kirk Johnson, Toshihiko Takeuchi, and Raphael Levy

XOMA Corporation, 2910 Seventh Street, Berkeley, CA 94710, United States

ABSTRACT

Humoral Hypercalcemia of Malignancy (HHM) occurs in 20-30 percent of advanced cancers, both solid tumors and hematologic malignancies, and is the most common life-threatening complication of cancer. HHM is due to tumors secreting abnormally high quantities of parathyroid hormone-related protein (PTHrP), a ligand for PTH1R (a family B GPCR) resulting in hypercalcemia. A potent and long acting PTH1R antagonist could reverse the hypercalcemia in HHM. Using XOMA's fully-human antibody phage display libraries, highly potent anti-PTH1R antagonist monoclonal antibodies were discovered, and screened by FACS for binding to CHO-K1 cells over-expressing human PTH1R. Human and murine receptor antagonism was demonstrated in cAMP accumulation and calcium flux assays with the human Saos-2 and the rat UMR106 osteosarcoma cell lines, which express endogenous PTH1R. The epitope demonstrating the greatest receptor antagonism was localized to the N-terminal extracellular domain of human PTH1R. Antibodies were affinity matured via light chain shuffling, resulting in leads with sub-nanomolar binding affinities as measured by SPR and verified by FACS. Functionally, PTH1R is expressed on osteoblasts and osteocytes. Stimulation with PTHrP leads to increased expression of M-CSF, RANKL and other factors that drive the differentiation and activation of bone resorbing osteoclast cells. It was shown that the lead anti-PTH1R mAb inhibited both PTH- and PTHrP-induced osteoclast differentiation by greater than 10-fold. In vivo proof-of-concept was achieved in rodent models where hypercalcemia was established in rats by SC infusion of PTHrP via an osmotic pump. IV administration of 2 and 10 mg/kg antibody reduced serum calcium levels by a minimum of 2 mg/dL within 48 hours in a dose-dependent manner. Additionally, in a murine C26 tumor model of hypercalcemia associated with elevated PTHrP, the anti-PTH1R mAb showed a dose-dependent correction of hypercalcemia at doses as low as 2 mg/kg with a sustained duration of action. In this model, the antibody given at 10 mg/kg IV completely reversed the hypercalcemia within 24 hours, lowering serum calcium from greater than 15 mg/dL to approximately 6mg/dL. Additional PK/PD parameters were assessed in mouse and rat models. Patients with HHM typically have a poor prognosis and HHM is a frequent reason that cancer patients are readmitted to hospitals or enter hospice care. Left untreated, hypercalcemia in these patients can cause coma or death. The highly potent anti-PTH1R antagonist mAb described here has the potential to become a first in class therapy for HHM, ameliorating hypercalcemia-associated morbidities, and extending the utility of other anti-cancer agents. This promising therapeutic antibody is currently in late stage preclinical development.

INTRODUCTION

Humoral Hypercalcemia of Malignancy (HHM) occurs in 20-30 percent of advanced cancers, both solid tumors and hematologic malignancies, and is the most common life-threatening complication of cancer. HHM is due to tumors secreting abnormally high quantities of parathyroid hormone-related protein (PTHrP), a ligand for PTH1R (a family B GPCR) resulting in hypercalcemia. A potent and long acting PTH1R antagonist could reverse the hypercalcemia in HHM. Patients with HHM typically have a poor prognosis and HHM is a frequent reason that cancer patients are readmitted to hospitals or enter hospice care. Left untreated, hypercalcemia in these patients can cause coma or death. The highly potent anti-PTH1R antagonist mAb described here has the potential to become a first in class therapy for HHM, ameliorating hypercalcemia-associated morbidities, and extending the utility of other anti-cancer agents. This promising therapeutic antibody is currently in late stage preclinical development.

- G-protein-coupled receptors (GPCRs) are broadly considered to be the most druggable class of therapeutic targets and play an important role in many indications where there is a high unmet medical need including cancer[1]
- Despite the value of targeting GPCRs, developing anti-GPCR antibodies has been a challenge due to their highly variable conformation and limited accessibility of GPCR functional epitopes[1]
- PTH1R is a B family G-Protein Coupled Receptor signaling via multiple pathways following activation with PTH or PTHrP ligands[2] (Figure 1)
- Inhibiting PTH1R signaling represents a potentially highly valuable and novel therapeutic mechanism of action for several hypercalcemic indications, such as HHM.

Fig 1

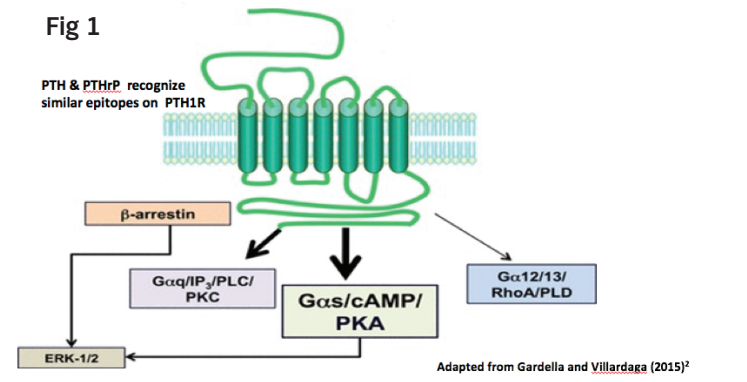

PTH & PTHrP recognize similar epitopes on PTH1R
β-arrestin
Gαq/IP3/PLC/PKC
Gαs/cAMP/PKA
Gα12/13/RhoA/PLD
ERK-1/2

Adapted from Gardella and Villardaga (2015)[2]

RESULTS: DISCOVERY AND CHARACTERIZATION IN VITRO

PTH1R-specific Monoclonal Antibodies Selected Using Phage Display Protocols

- XOMA's Fab and scFv phage display libraries[3,4] were panned on CHO-K1 cells overexpressing human PTH1R with N-terminal FLAG tag (CHO human PTH1R cells)
- XPA.85.012 & XPA.85.017 IgGs were chosen for further binding and functional characterization

Binding of XPA.85.012 and XPA.85.017 mAbs to Human PTH1R Expressed on CHO-K1 Cells

- **Flow cytometry screening of periplasmic extracts (Figure 2A,B):** Histogram plots for binding to CHO Human PTH1R (red) and CHO-K1 (blue) cells at a fixed concentration. Method: Periplasmic extracts tested for binding with anti-c-myc mAb, followed by anti-mouse IgG APC
- **Flow cytometry screening of reformatted IgGs (Figure 3A,B):** Binding curves to CHO Human PTH1R (red) and CHO-K1 (black) at increased antibody concentrations. Method: Reformatted IgGs tested for binding with anti-human IgG PE

Histogram Plots of Periplasmic Extract Clones

Binding Curves of Reformatted IgGs

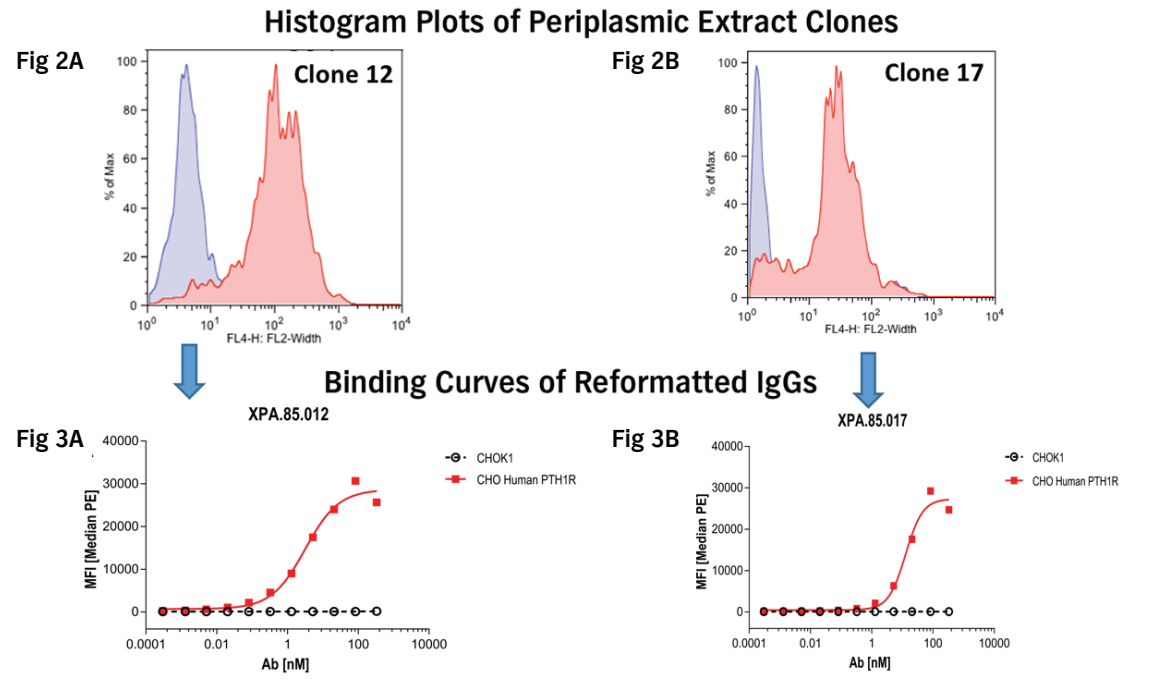

Fig 2A
Clone 12
Fig 2B
Clone 17
% of Max
FL4-H: FL2-Width
Fig 3A
XPA.85.012
Fig 3B
XPA.85.017
CHOK1
CHO Human PTH1R
MFI (Median PE)
Ab [nM]

XPA.85.012 and XPA.85.017 IgGs demonstrated specific binding to the human PTH1R receptor expressed on CHO-K1 cells

XPA.85.012 and XPA.85.017 cross-react to human and murine PTH1R in the presence or absence of PTH or PTHrP

- Increasing concentrations of anti-PTH1R antibodies incubated for 24 hr with cells expressing PTH1R orthologs in the presence or absence of saturating concentration of PTH or PTHrP
- Antibody binding detected by Flow Cytometry with R-Phycoerythrin anti-human IgG Abs. Binding to CHO-K1 cells expressing human PTH1R ± PTH (Figure 4A) or ± PTHrP (Figure 4B), or expressing mouse PTH1R ± PTH (Figure 4C) or ± PTHrP (Figure 4D)

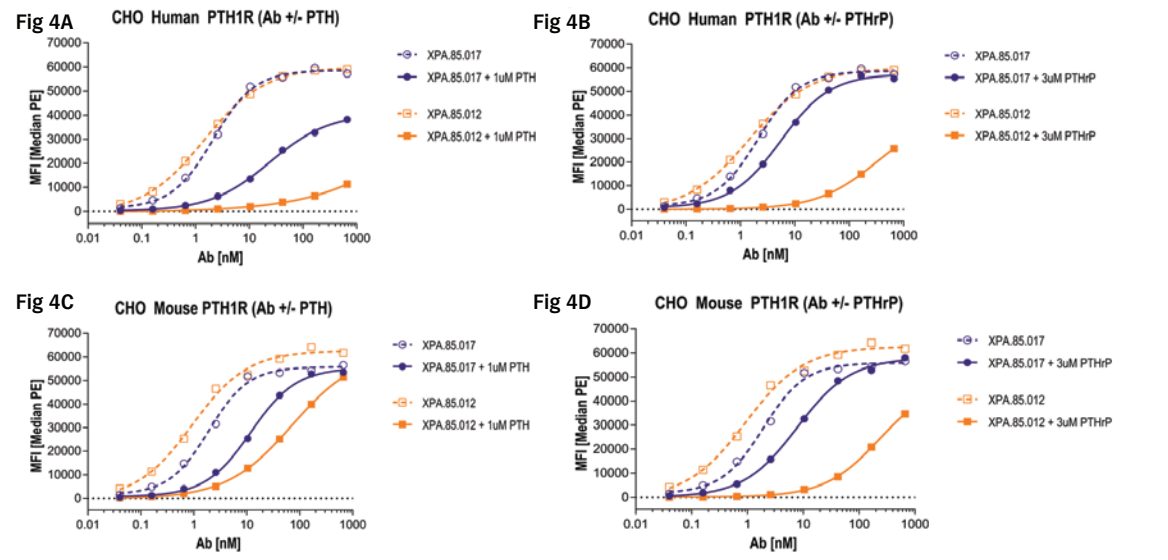

Fig 4A
CHO Human PTH1R (Ab +/- PTH)
Fig 4B
CHO Human PTH1R (Ab +/- PTHrP)
Fig 4C
CHO Mouse PTH1R (Ab +/- PTH)
Fig 4D
CHO Mouse PTH1R (Ab +/- PTHrP)
XPA.85.017
XPA.85.017 + 1uM PTH
XPA.85.012
XPA.85.012 + 1uM PTH
XPA.85.017 + 3uM PTHrP
XPA.85.012 + 3uM PTHrP
MFI (Median PE)
Ab [nM]

Both XPA.85.012 and XPA.85.017 cross-react with human and mouse PTH1R orthologs and both had increased binding in the absence of the PTH and PTHrP ligands (dotted lines) than in their presence (solid lines)

XPA.85.012 and XPA.85.017 Inhibit Ligand-Mediated cAMP Accumulation and Calcium Mobilization

- CHO human PTH1R cells were preincubated for 30 min at 37°C with 40 µg/ml of XPA.85.012 (red triangles) or XPA.85.017 (blue squares).
- Induction performed with increasing concentrations of PTH (1-34) (Figure 5A,6C) or PTHrP (Figure 5B,6D)

cAMP accumulation

Ca^{+2} Mobilization

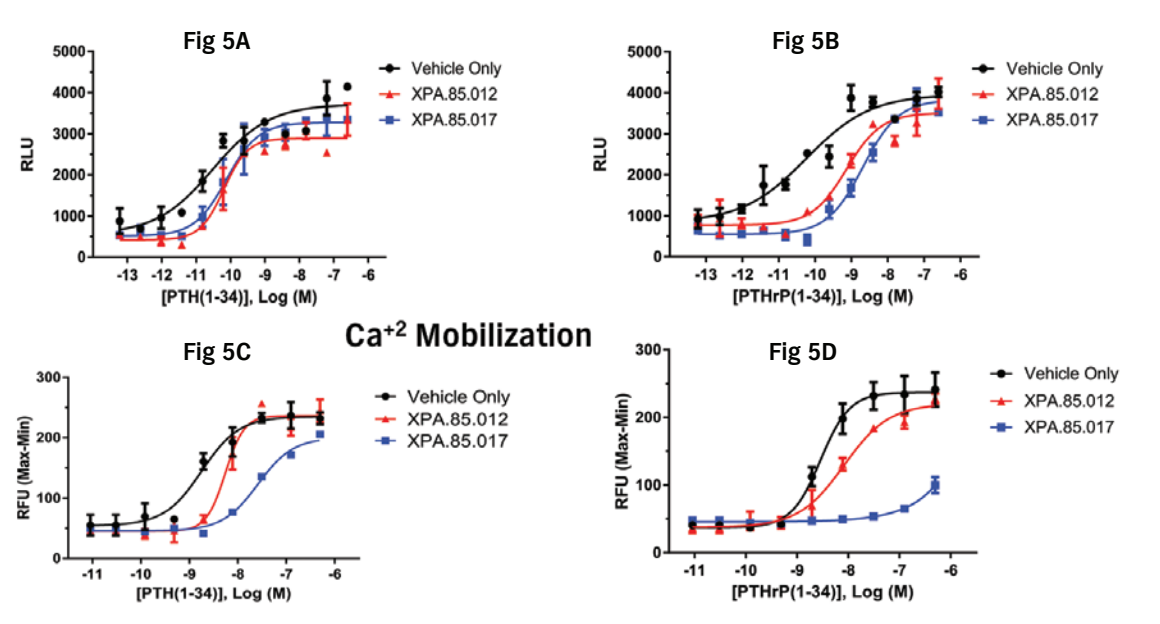

Fig 5A
Fig 5B
Fig 5C
Fig 5D
Vehicle Only
XPA.85.012
XPA.85.017
RLU
RFU (Max-Min)
[PTH(1-34)], Log (M)
[PTHrP(1-34)], Log (M)

Both XPA.85.012 and XPA.85.017 inhibited PTH and PTHrP-driven cAMP accumulation (Gs/PKA pathway, Figure 5A, 5B) and calcium mobilization (Gq/PKC pathway, Figure 5C, 5D)

The XPA.85.012 and XPA.85.017 Epitopes are Located in the PTH1R N-Terminal Extracellular Domain (N-ECD)

- XPA.85.012 & XPA.85.017 do not recognize human N-terminal-deleted PTH1R (NTD) on CHO-K1 cells (Figure 6A, 6B). Method: Antibodies were incubated with CHO-human PTH1R or CHO-truncated human PTH1R (NTD) for 30 min and then detected with conjugated anti-human-Fc-specific antisera

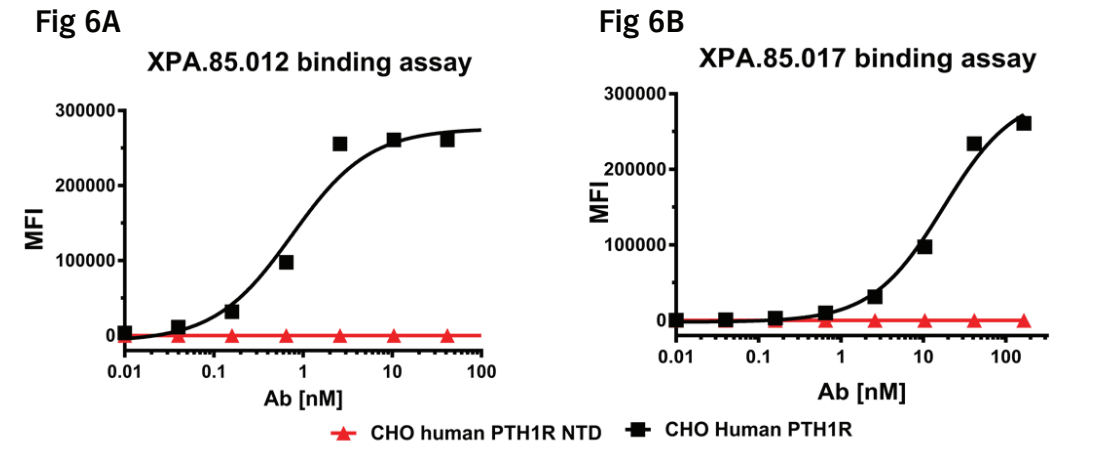

Fig 6A
XPA.85.012 binding assay
Fig 6B
XPA.85.017 binding assay
MFI
Ab [nM]
CHO human PTH1R NTD
CHO Human PTH1R

- XPA.85.012 & XPA.85.017 recognize a commercially available soluble recombinant N-ECD by SPR (Figure 7A, 7B). Method: Antibodies were captured via an anti-Fc monoclonal on a Biacore 2000. PTH1R N-Terminus (N-ECD) was injected at 150 nM, 30 nM, 6 nM, and 1.2 nM

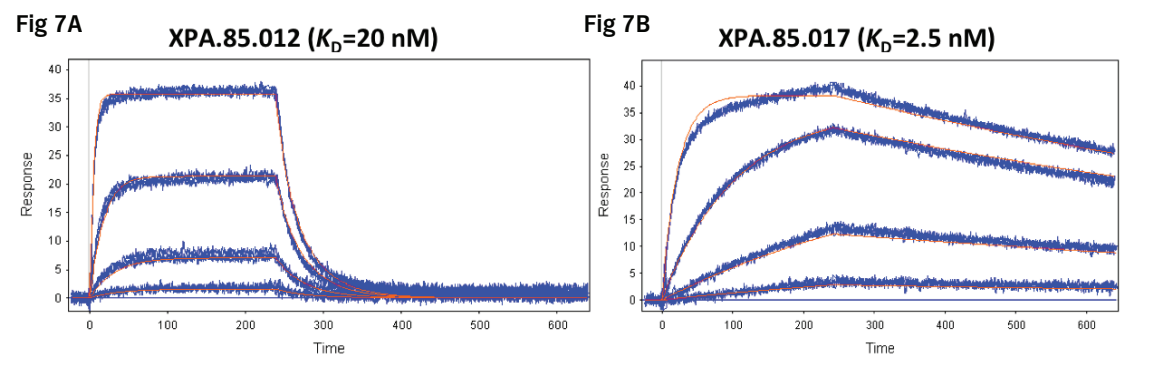

Fig 7A
XPA.85.012 (KD=20 nM)
Fig 7B
XPA.85.017 (KD=2.5 nM)
Response
Time

Subsequently, both XPA.85.012 and XPA.85.017 IgGs were engineered via light chain shuffling to improve their binding affinity

Affinity Maturation of Anti-PTH1R XPA.85.012 & XPA.85.017 via Light Chain Shuffling

- Three rounds of soluble phage panning were performed vs recombinant human N-ECD of PTH1R with increasing stringency of selection as XPA.85.012 & XPA.85.017 only recognize full-length PTH1R on CHO-K1 cells and not the N-terminally deleted human PTH1R (Figure 9)

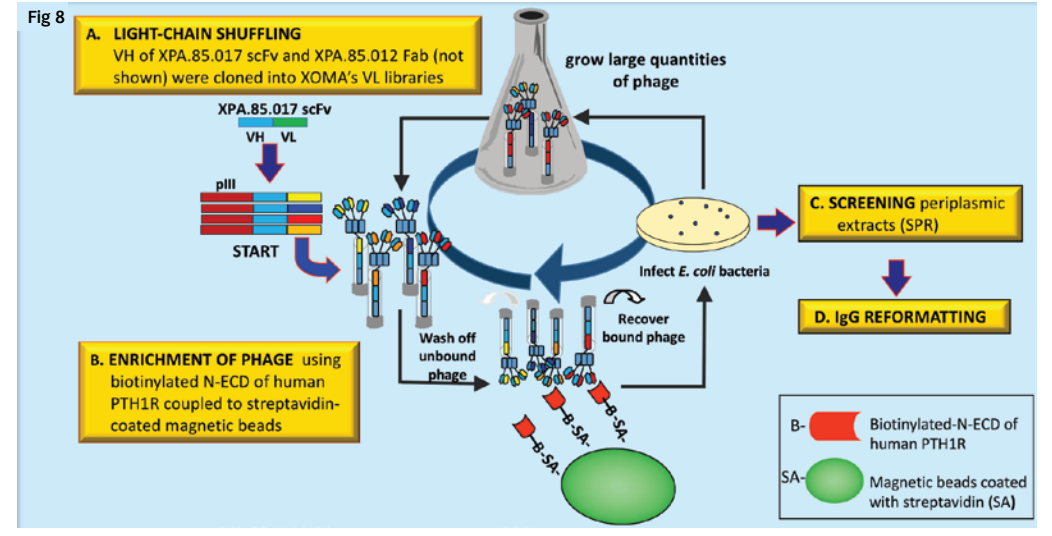

Fig 8
A. LIGHT-CHAIN SHUFFLING
VH of XPA.85.017 scFv and XPA.85.012 Fab (not shown) were cloned into XOMA's VL libraries
XPA.85.017 scFv
VH VL
pIII
START
grow large quantities of phage
C. SCREENING periplasmic extracts (SPR)
D. IgG REFORMATTING
Infect E. coli bacteria
Recover bound phage
Wash off unbound phage
B. ENRICHMENT OF PHAGE using biotinylated N-ECD of human PTH1R coupled to streptavidin-coated magnetic beads
Biotinylated-N-ECD of human PTH1R
Magnetic beads coated with streptavidin (SA)

Representative SPR-derived Kinetic Data Comparing Parental XPA.85.012 and XPA.85.017 IgGs with their Respective Higher Affinity Light Chain Variants

- IgGs were captured to the SPR sensor surface via the Fc region and recombinant human PTH1R N-terminal extracellular domain (N-ECD) was injected at 200, 50, 12.5, and 3.125 nM (Figure 9)

Table 1. SPR Based Affinity Estimates

Clone	ka (1/Ms)	kd (1/s)	K_D (M)
XPA.85.012	1.3E+06	2.5E-02	2.0E-08
XPA.85.017	3.5E+05	8.6E-04	2.5E-09
XPA.85.287	3.6E+06	3.6E-04	9.9E-11
XPA.85.288	7.8E+05	1.4E-04	1.8E-10

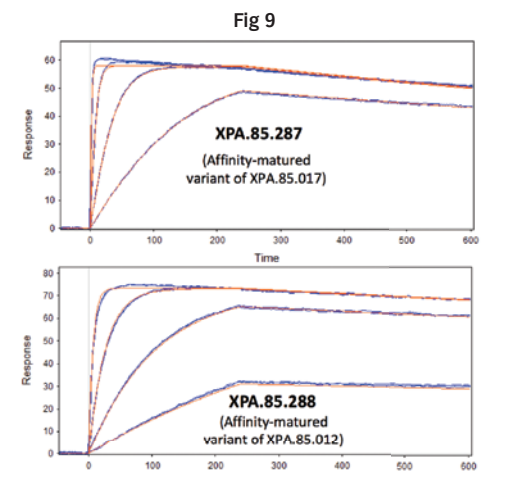

Fig 9
XPA.85.287
(Affinity-matured variant of XPA.85.017)
XPA.85.288
(Affinity-matured variant of XPA.85.012)
Response
Time

Affinity matured XPA.85.012 and XPA.85.017 variants shown to have subnanomolar K_Ds

XOMA's XPA.85.287 and XPA.85.288 Antagonist Abs Behave as Negative Allosteric Modulators of the PTH1R Receptor

- Concentration-response curves for PTHrP(1-34) and increasing concentrations of XPA.85.287 mAb in cAMP assays using CHO human PTH1R (Figure 10A) and CHO mouse PTH1R (Figure 10B) cells
- Concentration-response curves for PTHrP (1-34) and increasing concentrations of XPA.85.288 mAb in cAMP assays using CHO human PTH1R (Figure 10C) and CHO mouse PTH1R (Figure 10D)

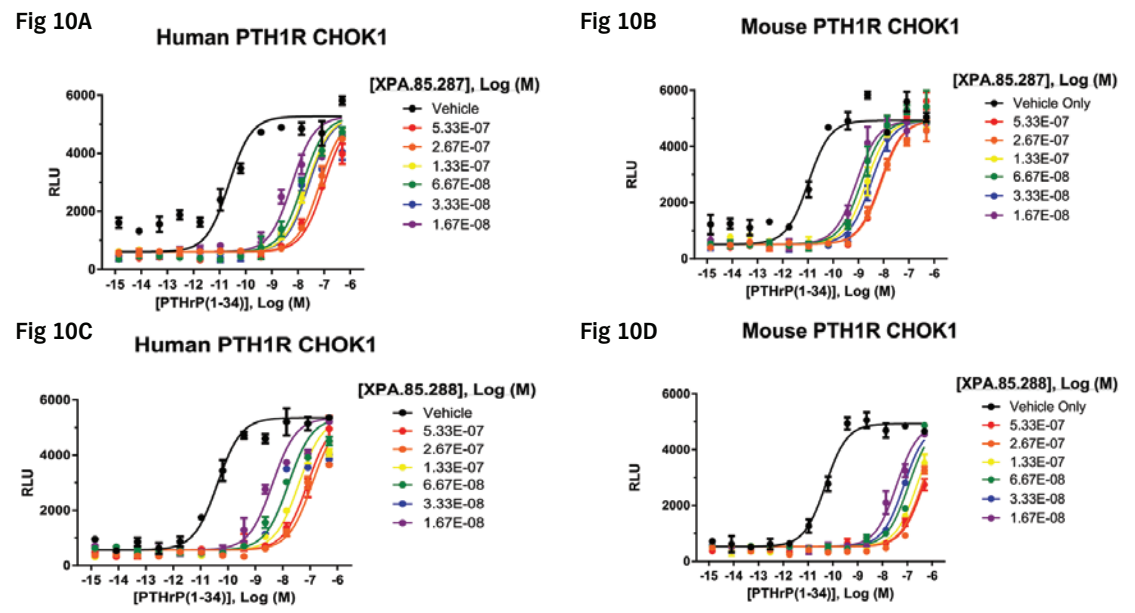

Fig 10A
Human PTH1R CHOK1
Fig 10B
Mouse PTH1R CHOK1
Fig 10C
Human PTH1R CHOK1
Fig 10D
Mouse PTH1R CHOK1
[XPA.85.287], Log (M)
[XPA.85.288], Log (M)
Vehicle
Vehicle Only
5.33E-07
2.67E-07
1.33E-07
6.67E-08
3.33E-08
1.67E-08
RLU
[PTHrP(1-34)], Log (M)

Schild regression analyses (GraphPad Software Inc, CA) revealed non-parallel rightward shifts in concentration-response curves, consistent with non-competitive (allosteric) mechanism of action of both XPA.85.287 and XPA.85.288

Functional XPA.85.288, XPA.85.287 Activities in cAMP Assays Using Endogenous Cell Lines Expressing Rat or Human PTH1R

- IgGs at 40 µg/ml were preincubated with human SaoS-2 (Figure 11A, B) or rat UMR106 (Figure 11C) cells for 30 min at 37°C prior to induction of cAMP accumulation by PTH (1-34) for 45 min at 37°C

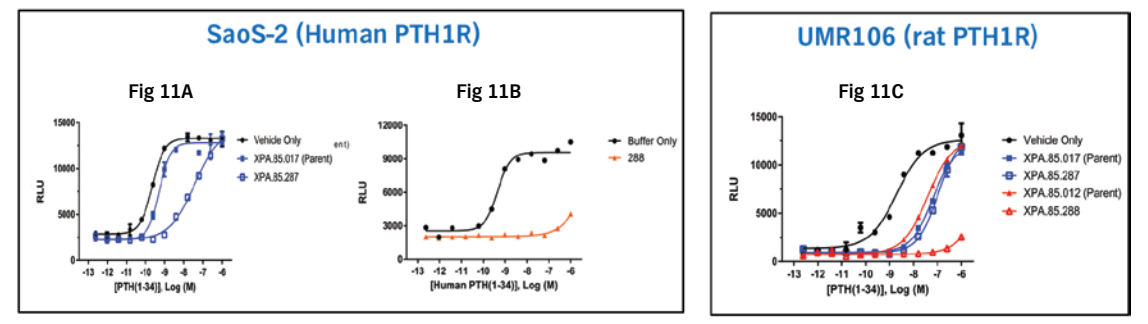

SaoS-2 (Human PTH1R)
Fig 11A
Fig 11B
UMR106 (rat PTH1R)
Fig 11C
Vehicle Only
XPA.85.017 (Parent)
XPA.85.287
Buffer Only
288
XPA.85.012 (Parent)
XPA.85.288
RLU
[PTH(1-34)], Log (M)
[Human PTH(1-34)], Log (M)

- *Following PTH activation, affinity-maturated IgG XPA.85.287 showed significantly improved activity against native human PTH1R vs parent XPA.85.017 (Figure 11A), with little change in activity against native rat PTH1R (Figure 11C)*
- *In contrast, following PTH activation, affinity-maturated IgG XPA.85.288 showed significantly improved activity against both native human PTH1R (Figure 11B) and native rat PTH1R (Figure 11C) vs parent XPA.85.012 (data not shown)*

Effects of PTH1R XPA.85.288 and XPA.85.287 IgGs on PTH and PTHrP-stimulated M-CSF Secretion by SaoS-2 Osteosarcoma Cells

Rationale: *PTH & PTHrP act on PTH1R expressed in osteoblasts and osteocytes (i.e. SaoS-2 cells) to stimulate production of macrophage colony stimulating factor (M-CSF). Increased production of M-CSF drives differentiation and activation of bone resorbing osteoclasts, leading to increased level of calcium efflux from bone and bone demineralization. M-CSF is required for PTH and PTHrP-mediated differentiation and activation of human osteoclasts.[5]*

- Saos-2 cells were stimulated for 48h with PTH (1-34) (Figure 12A) or PTHrP (1-36) (Figure 12B) in the presence of 40 µg/ml antibody. XPA.85.012 (green square), XPA.85.017 (purple triangle), XPA.85.287 (red inverted triangle), XPA.85.288 (blue diamond) , negative control IgG (black circle). M-CSF secretion by Saos-2 cells was measured by immunoassay and read on a Sector Imager 6000.

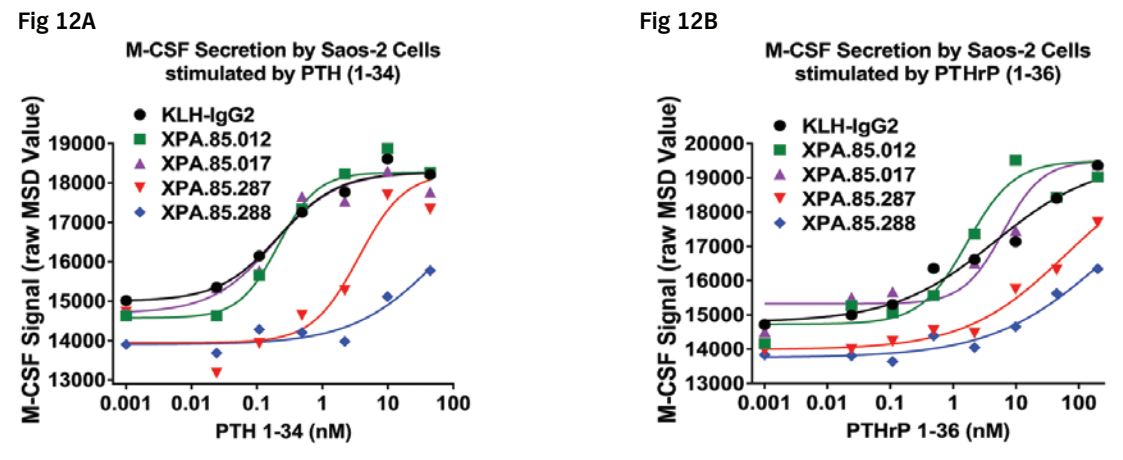

Fig 12A
M-CSF Secretion by Saos-2 Cells stimulated by PTH (1-34)
Fig 12B
M-CSF Secretion by Saos-2 Cells stimulated by PTHrP (1-36)
KLH-IgG2
XPA.85.012
XPA.85.017
XPA.85.287
XPA.85.288
M-CSF Signal (raw MSD Value)
PTH 1-34 (nM)
PTHrP 1-36 (nM)

Affinity-matured anti-PTH1R mAbs XPA.85.288 and XPA.85.287 were shown to inhibit PTH and PTHrP-stimulated M-CSF secretion

RESULTS: CHARACTERIZATION IN VIVO

Anti-PTH1R XPA.85.288 mAb Reduced PTH-induced Elevation of Serum Calcium Levels in a Thyroparathyroidectomized Rat Model

Rationale: *Thyroparathyroidectomized (TPTx) rats with hypocalcemia were used to test the ability of anti-PTH1R antibodies to reverse serum calcium level increases upon infusion of PTH.*

Study protocol:

- Sprague-Dawley male rats (n=5-6/group) challenged intravenously with anti-PTH1R mAbs XPA.85.017, XPA.85.287, XPA.85.288, anti-PTH control (XPA.85.BM2), isotype control (15 mg/kg), 18 hr before initiation of PTH(1-34) infusion.
- Serum calcium was measured before dosing (Predose baseline), Pre infusion, 2, 4, 6 hr post start infusion (Figure 13)

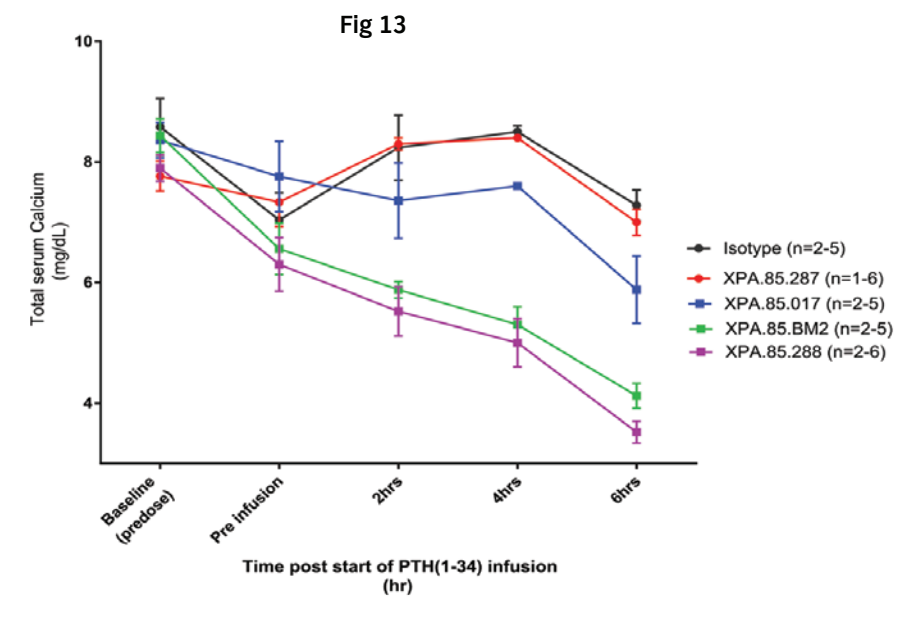

Fig 13
Total serum Calcium (mg/dL)
Isotype (n=2-5)
XPA.85.287 (n=1-6)
XPA.85.017 (n=2-5)
XPA.85.BM2 (n=2-5)
XPA.85.288 (n=2-6)
Baseline (predose)
Pre infusion
2hrs
4hrs
6hrs
Time post start of PTH(1-34) infusion (hr)

XPA.85.288 significantly reduced PTH-induced increase of serum calcium in TPTx rats thus normalizing serum calcium

Efficacy of Anti-PTH1R mAb on Mouse Colon 26 Tumor-related Hypercalcemia

Rationale: *Evaluation of serum calcium and albuminin in the subcutaneously (SC)-implanted Colon 26 murine colon tumor model treated with XOMA antibody XPA.85.349.*

Study protocol:

- CDF1 mice female (n=3-8/group) challenged subcutaneously with Colon 26 murine colon tumor cell line at 7.5X10^6 cells/each
- Treatment initiated when tumors size greater than 1000 mg
- Anti-PTH1R mAb administered 6 and 2 mg/kg intravenously. Tumor weight and serum samples were collected at pretreatment, 24, 48 and 72 hours post-treatment.
- Calcium level were collected at no implant, pretreatment, 24, 48, 72 and 120 hours post-treatment.

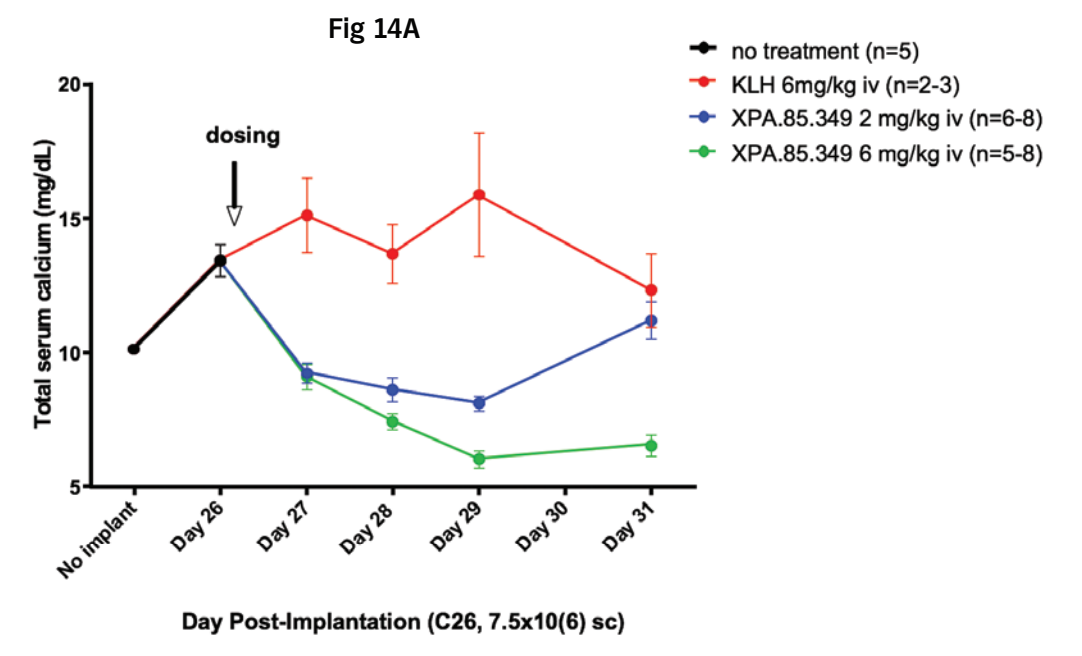

Fig 14A
no treatment (n=5)
KLH 6mg/kg iv (n=2-3)
XPA.85.349 2 mg/kg iv (n=6-8)
XPA.85.349 6 mg/kg iv (n=5-8)
dosing
Total serum calcium (mg/dL)
No implant
Day 26
Day 27
Day 28
Day 29
Day 30
Day 31
Day Post-Implantation (C26, 7.5x10(6) sc)

Anti-PTH1R mAb significantly reduced C26-induced serum calcium in CDF1 mice

Body Weight, Tumor Weight, and Serum PTHrP and PTH

- Body weight and tumor size were measured at -24 hours, 48, 72 and 120 hours post-treatment.
- Levels of PTHrP and PTH in serum at different time points post dosing. Antibodies were administered intraveneously at 2 and 6 mg/kg at t0 (n=2-6 mice/group). Blood samples were collected from retro-orbital sinus at 24, 48, 72 and 120 hours post administration and the concentrations of PTHrP (Phoenix Pharmaceuticcals, Inc - EIA Kit, cat#: EK-056-04) and mouse PTH 1-84 (QUIDEL Mouse PTH 1-84 ELISA kit, cat#: 60-2305) measured.

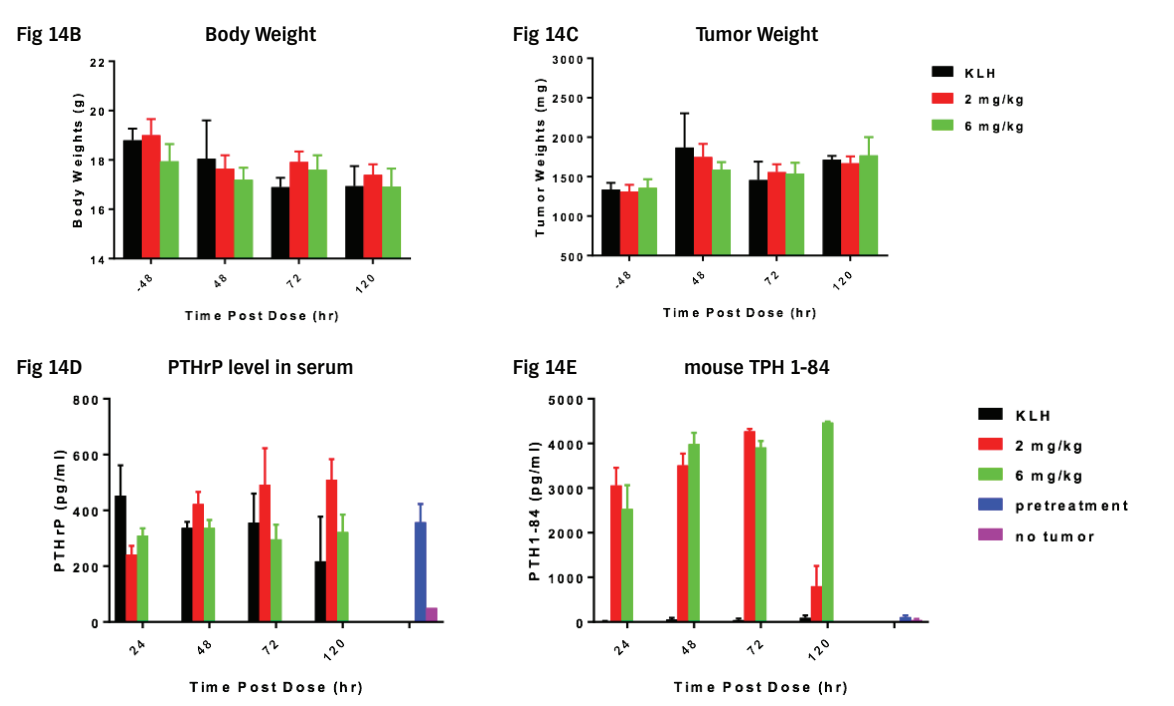

Fig 14B
Body Weight
Fig 14C
Tumor Weight
Fig 14D
PTHrP level in serum
Fig 14E
mouse TPH 1-84
KLH
2 mg/kg
6 mg/kg
pretreatment
no tumor
Body Weights (g)
Tumor Weights (mg)
PTHrP (pg/ml)
PTH1-84 (pg/ml)
Time Post Dose (hr)

Mice treated with anti-PTH1R mAb showed no significant reduction in body or tumor weight up to 120 hours post-treatment

Tumor implantation increased serum PTHrP levels which were unchanged 120 hours post treatment with anti-PTH1R mAb. In constrast, serum PTH levels were significantly increased following treatment.

PK of XPA.85.349 in mice

- PK profile of XPA.85.349 via intravenous injection. CDF1 mice female, administered as single dose bolus at 2 and 6 mg/kg. Blood samples were collected from retro-orbital sinus at 24, 48, 72 and 120 hours after the administration of XPA.85.349 and its concentration measured by ELISA. Data were analyzed using WinNonlin.

Table 2.

	2 mg/kg	6 mg/kg
Co (µg/mL)	7.2	47.8
AUC_{last} (µg*hr/mL)	414	1832
AUC_{inf} (µg*hr/mL)	438	2116
$t_{1/2}$ (hr)	26	48
Vz (mL/kg)	172	195
Cl (mL/hr/kg)	4.6	2.8

Anti-PTH1R mAb XPA.85.349 has a half-life of up to 48hrs in mice.

CONCLUSIONS

- **XOMA's antibody libraries enabled the discovery of functional antibodies against the very complex G-Protein Coupled Receptor (PTH1R)**
- **Lead anti-PTH1R mAbs:**
 - Antagonized PTH1R, reducing ligand-induced cAMP production and calcium mobilization
 - Cross-reacted with human and mouse PTH1R orthologs
 - Recognized epitopes within the N-terminal extracellular domain (N-ECD) of human PTH1R
- **Affinity maturation resulted in antibodies with improved kinetic and functional properties:**
 - Both XPA.85.288 and XPA.85.287 variants showed increased receptor antagonism
 - XPA.85.288 abrogated PTH1R activity and inhibited M-CSF secretion in a model of hypercalcemia using endogenous osteosarcoma cell lines
- **PTH1R antagonism in vitro by XPA.85.288 translated to potency in vivo**
 - Effect on Hypocalcemic TPTx rats treated with PTH to increase their serum calcium levels
 - Reduced hypercalcemia in HHM model with Colon 26 tumors
- **Anti-PTH1R antagonist mAb described has the potential to become a first in class therapy for HHM, ameliorating hypercalcemia-associated morbidities, and extending the utility of other anti-cancer agents.**

References

1. Migyeong Jo, and Sang Taek Jung., 2016, Engineering therapeutic antibodies targeting G-protein-coupled receptors. *Experimental & Molecular Medicine* 48, 1-9.
2. Gardella, T.J, and Vilardaga, J.P., 2015, International Union of Basic and Clinical Pharmacology. XCIII. The parathyroid hormone receptors–family B G protein-coupled receptors. *Pharmacol. Rev.* 67, 310-37.
3. Levy, R. et al., 2013, Enhancement of antibody fragment secretion into the Escherichia coli periplasm by co-expression with the peptidyl prolyl isomerase, FkpA, in the cytoplasm. *J. Immunol. Methods* 394, 10-21.
4. Schwimmer L.J., et al., 2013, Discovery of diverse and functional antibodies from large human repertoire antibody libraries. *J. Immunol. Methods* 391, 60-71.
5. Suda Tatsuo, et al., 1999, Modulation of Osteoclast Differentiation and Function by the New Members of the Tumor Necrosis Factor Receptor and Ligand Families. *Endocrine Reviews* 20, 345-57

XOMA